ASCO TAPUR
Targeted Agent and Profiling Utilization Registry Study

Abstract 106: Talazoparib in patients with advanced colorectal cancer with *BRCA1/2* mutations: Results from the Targeted Agent and Profiling Utilization Registry (TAPUR) Study

ASCO
AMERICAN SOCIETY OF CLINICAL ONCOLOGY

KJ Yost[1], M Rothe[2], PK Mangat[2], E Garrett-Mayer[2], HL Duvivier[3], ER Ahn[4], TL Cannon[5], VK Chiu[6], MF Khalil[7], BK Kim[8], A Prinz[9], GN Grantham[2], A Gregory[2], DC Hinshaw[2], S Halabi[10], RL Schilsky[2]
[1]Cancer Research Consortium of West Michigan, Grand Rapids, MI; [2]American Society of Clinical Oncology, Alexandria, VA; [3]City of Hope Atlanta, Newnan, GA; [4]City of Hope Chicago, Zion, IL; [5]Inova Schar Cancer Institute, Fairfax, VA; [6]The Angeles Clinic and Research Institute, A Cedars-Sinai Affiliate, Los Angeles, CA; [7]Lehigh Valley Topper Cancer Institute, Allentown, PA; [8]Sutter Sacramento Medical Center, Sacramento, CA; [9]Levine Cancer Institute, Atrium Health, Lincolnton, NC; [10]Duke University Medical Center, Durham, NC

Background:

- TAPUR is a phase II basket study that evaluates the antitumor activity of commercially available targeted agents in patients (pts) with advanced cancers with specific genomic alterations.
- **Results of a cohort of pts with colorectal cancer (CRC) with *BRCA1/2* mutations (mut) treated with talazoparib (tala) are reported.**

Methods:

Study Design:

- **Eligible pts:** Advanced CRC, ECOG performance status (PS) 0-2, adequate organ function, measurable disease, and no standard treatment options available. Treatment was assigned according to pre-specified matching rules based on genomic tests performed in CLIA-certified, CAP-accredited labs selected by sites.
- Pts received 1 mg of tala orally once daily until disease progression, unacceptable toxicity or pt or physician choice to discontinue.
- **Primary endpoint:** Disease control (DC) defined by investigator assessment of objective response (OR) or stable disease (SD) of at least 16+ weeks (wks) duration per RECIST v1.1. Radiographic confirmation of response was not required.
- **Secondary endpoints:** OR, progression-free survival (PFS), overall survival (OS), and toxicity per CTCAE v 4.0 are reported. For toxicity, grade 3-5 adverse events (AEs) or serious adverse events (SAEs) at least possibly related to tala are reported.

Statistical Methods:

- Simon's optimal two-stage design was used to test null hypothesis of 15% DC rate. Power of 85% and 1-sided α of 10% are based on alternative DC rate of 35%.
- If ≥2 of 10 pts in stage 1 have DC, 18 more pts are enrolled; otherwise, the cohort is closed. At least 7 of 28 pts at end of stage 2 must achieve DC to reject the null hypothesis and consider treatment worthy of further study.

Results:

- 10 pts were enrolled to stage 1 between February 2020 to June 2021. Pt demographics and clinical characteristics are outlined in **Table 1.**
- **Alterations**: 3 pts (30%) had *BRCA1* mut, 5 pt (50%) had *BRCA2* mut, and 2 pts (20%) had both *BRCA1/2*. For most pts, the genomic test performed did not distinguish between germline and somatic muts.
- **Outcomes**: All pts were evaluable for efficacy. No pts experienced OR or SD16+, yielding a DC rate of 0% (p=1.00); the null hypothesis was not rejected.

Conclusion: Talazoparib did not demonstrate sufficient clinical activity in heavily pre-treated patients with CRC with *BRCA1/2* mutations for continued evaluation in this patient population.

Future Direction: Other treatments should be considered for these patients, including treatments offered in clinical trials.

- **Safety:** 2 pts (20%) had ≥1 grade 3 AE at least possibly related to treatment, including anemia and fatigue.

Table 1. Clinical Characteristics (N=10)

Characteristic		No. (%)
Age (Years)	Median (range)	59 (51-85)
Sex	Female	5 (50)
Race	White	10 (100)
Ethnicity	Not Hispanic or Latino	10 (100)
ECOG PS	0 1 2	4 (40) 4 (40) 2 (20)
Prior Systemic Regimens	2 ≥3	2 (20) 8 (80)
Tumor Sidedness	Left Right Rectal Undetermined	2 (20) 2 (20) 3 (30) 3 (30)

Funding provided by Pfizer. The authors would like to thank the patients who participated in this cohort, the clinical centers and staff, as well as Betty "B" Thompson of Pfizer, a TAPUR supporting pharmaceutical company.
Contact: TAPURPublications@asco.org

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Table 2: Efficacy Outcomes (N=10)

DC rate, % (1-sided 90% CI) (p-value)	0 (0, 100) (p=1.00)
OR rate, % (95% CI)	0 (0, 31)
Median PFS, wks (95% CI)	8 (6, 8)
Median OS, wks (95% CI)	24 (7, 43)

Figure 1: Best Percent Change from Baseline in Target Lesion Size (N=10)

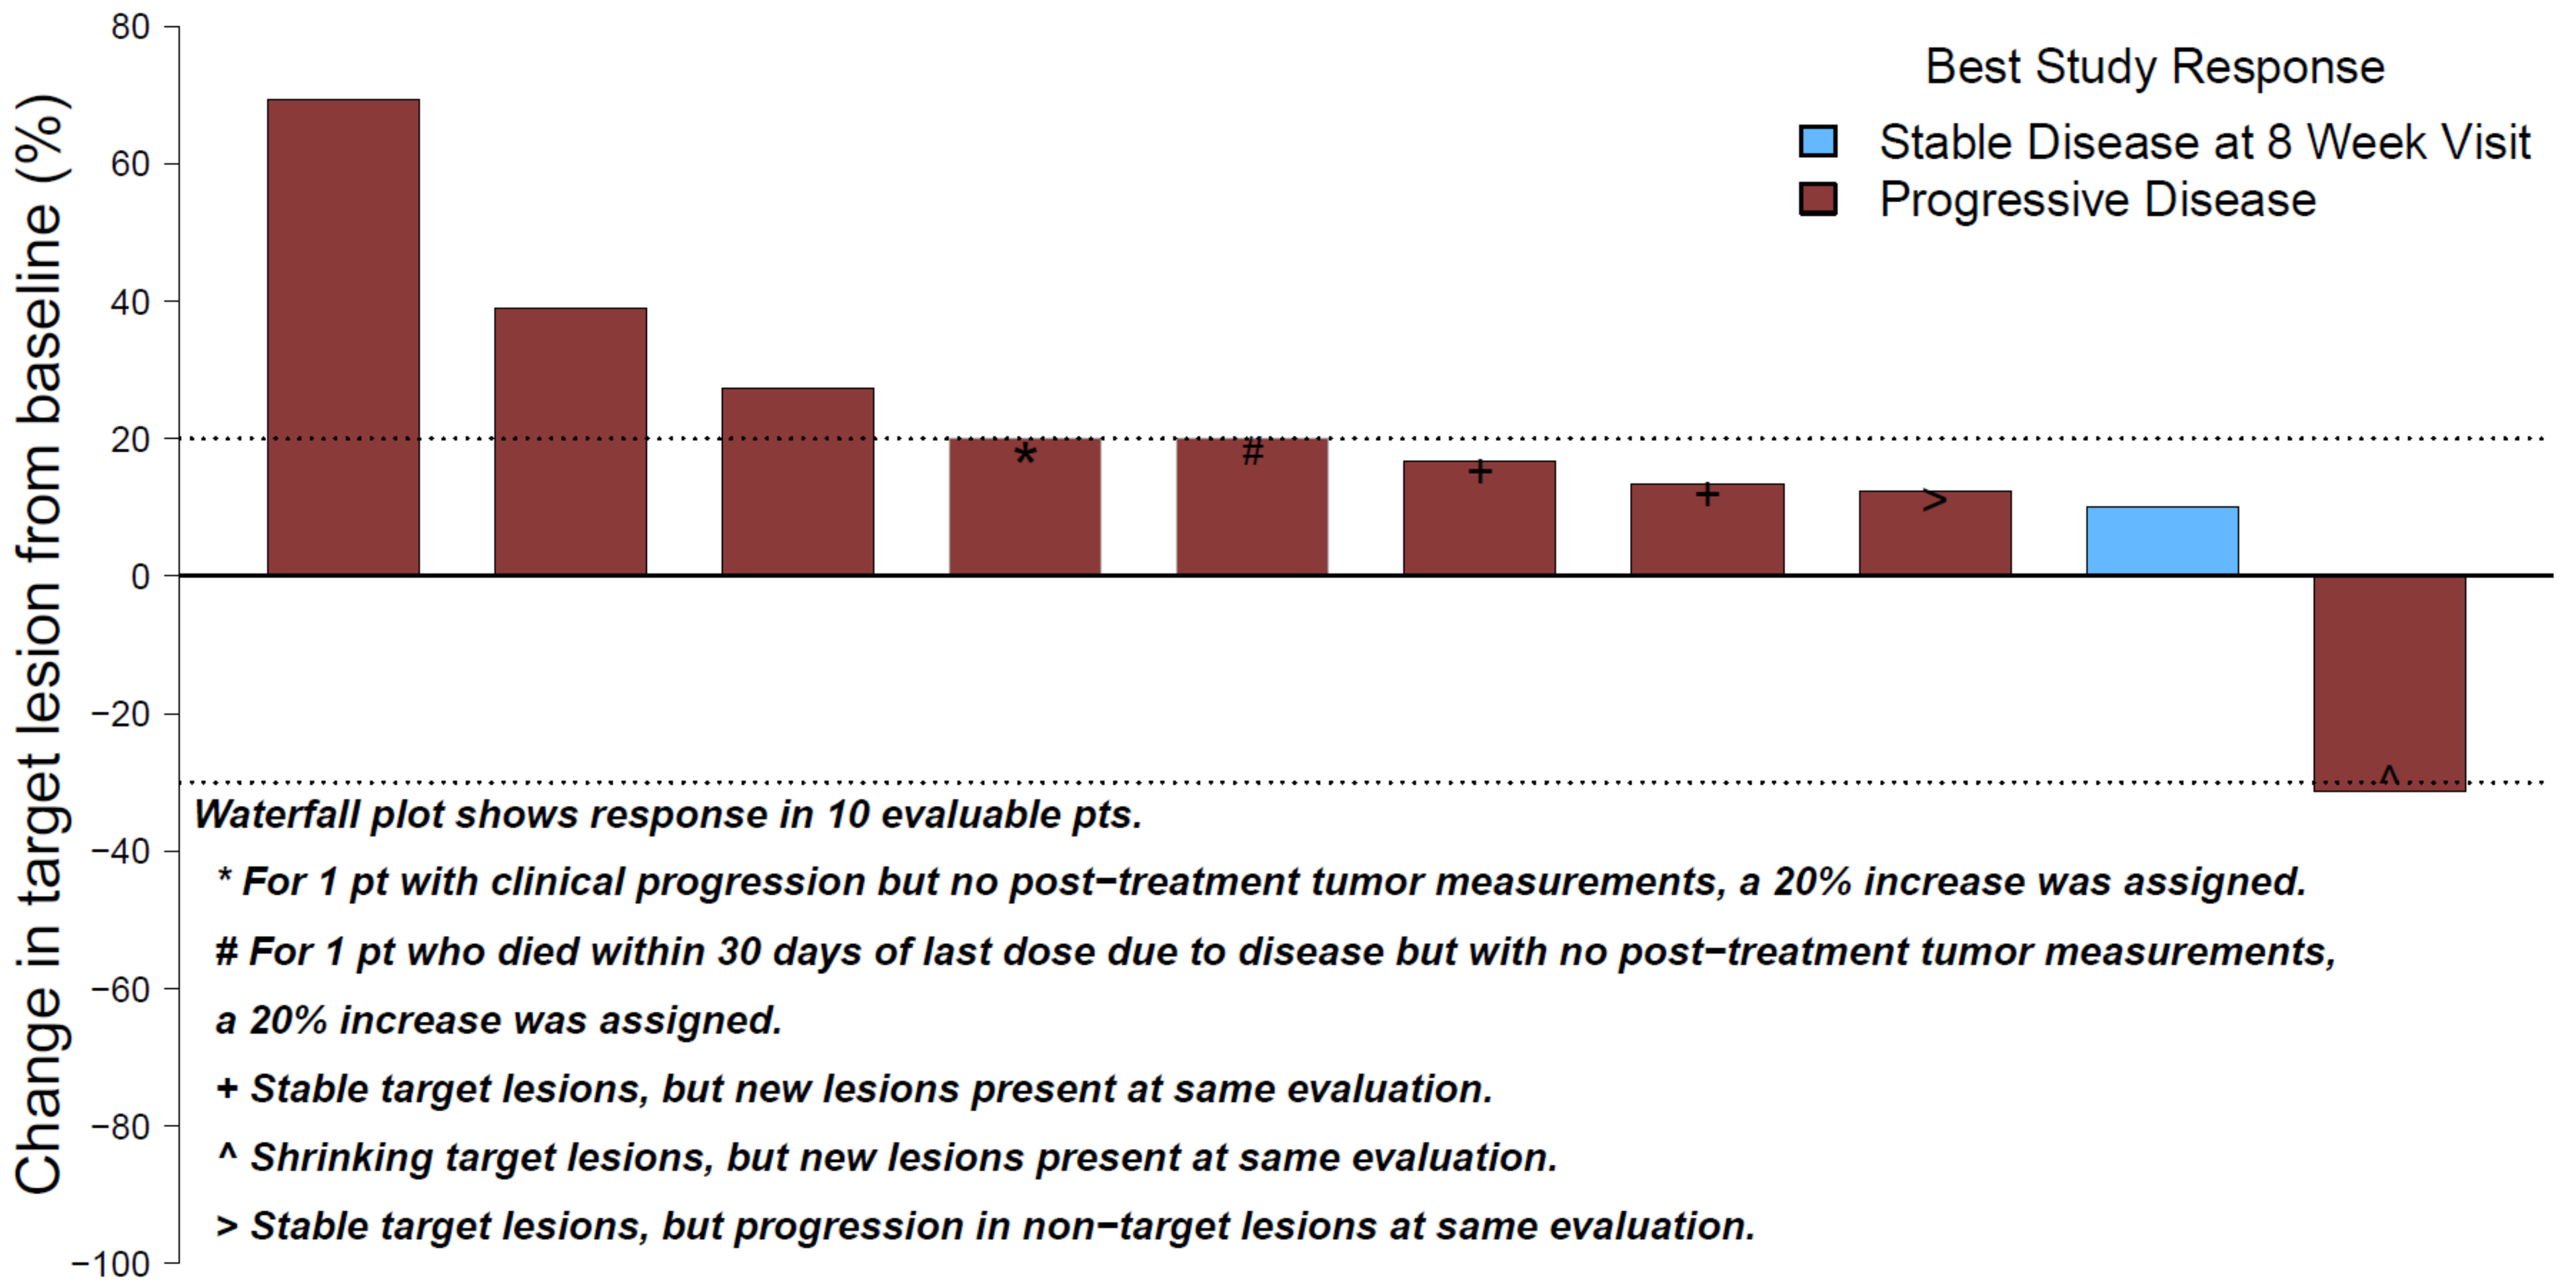
80
60
40
20
0
−20
−40
−60
−80
−100
Change in target lesion from baseline (%)
Best Study Response
Stable Disease at 8 Week Visit
Progressive Disease
*
#
+
+
>
^
Waterfall plot shows response in 10 evaluable pts.
* For 1 pt with clinical progression but no post−treatment tumor measurements, a 20% increase was assigned.
For 1 pt who died within 30 days of last dose due to disease but with no post−treatment tumor measurements, a 20% increase was assigned.
+ Stable target lesions, but new lesions present at same evaluation.
^ Shrinking target lesions, but new lesions present at same evaluation.
> Stable target lesions, but progression in non−target lesions at same evaluation.

Figure 2: Percent Change of Tumor Burden During Treatment (N=10)

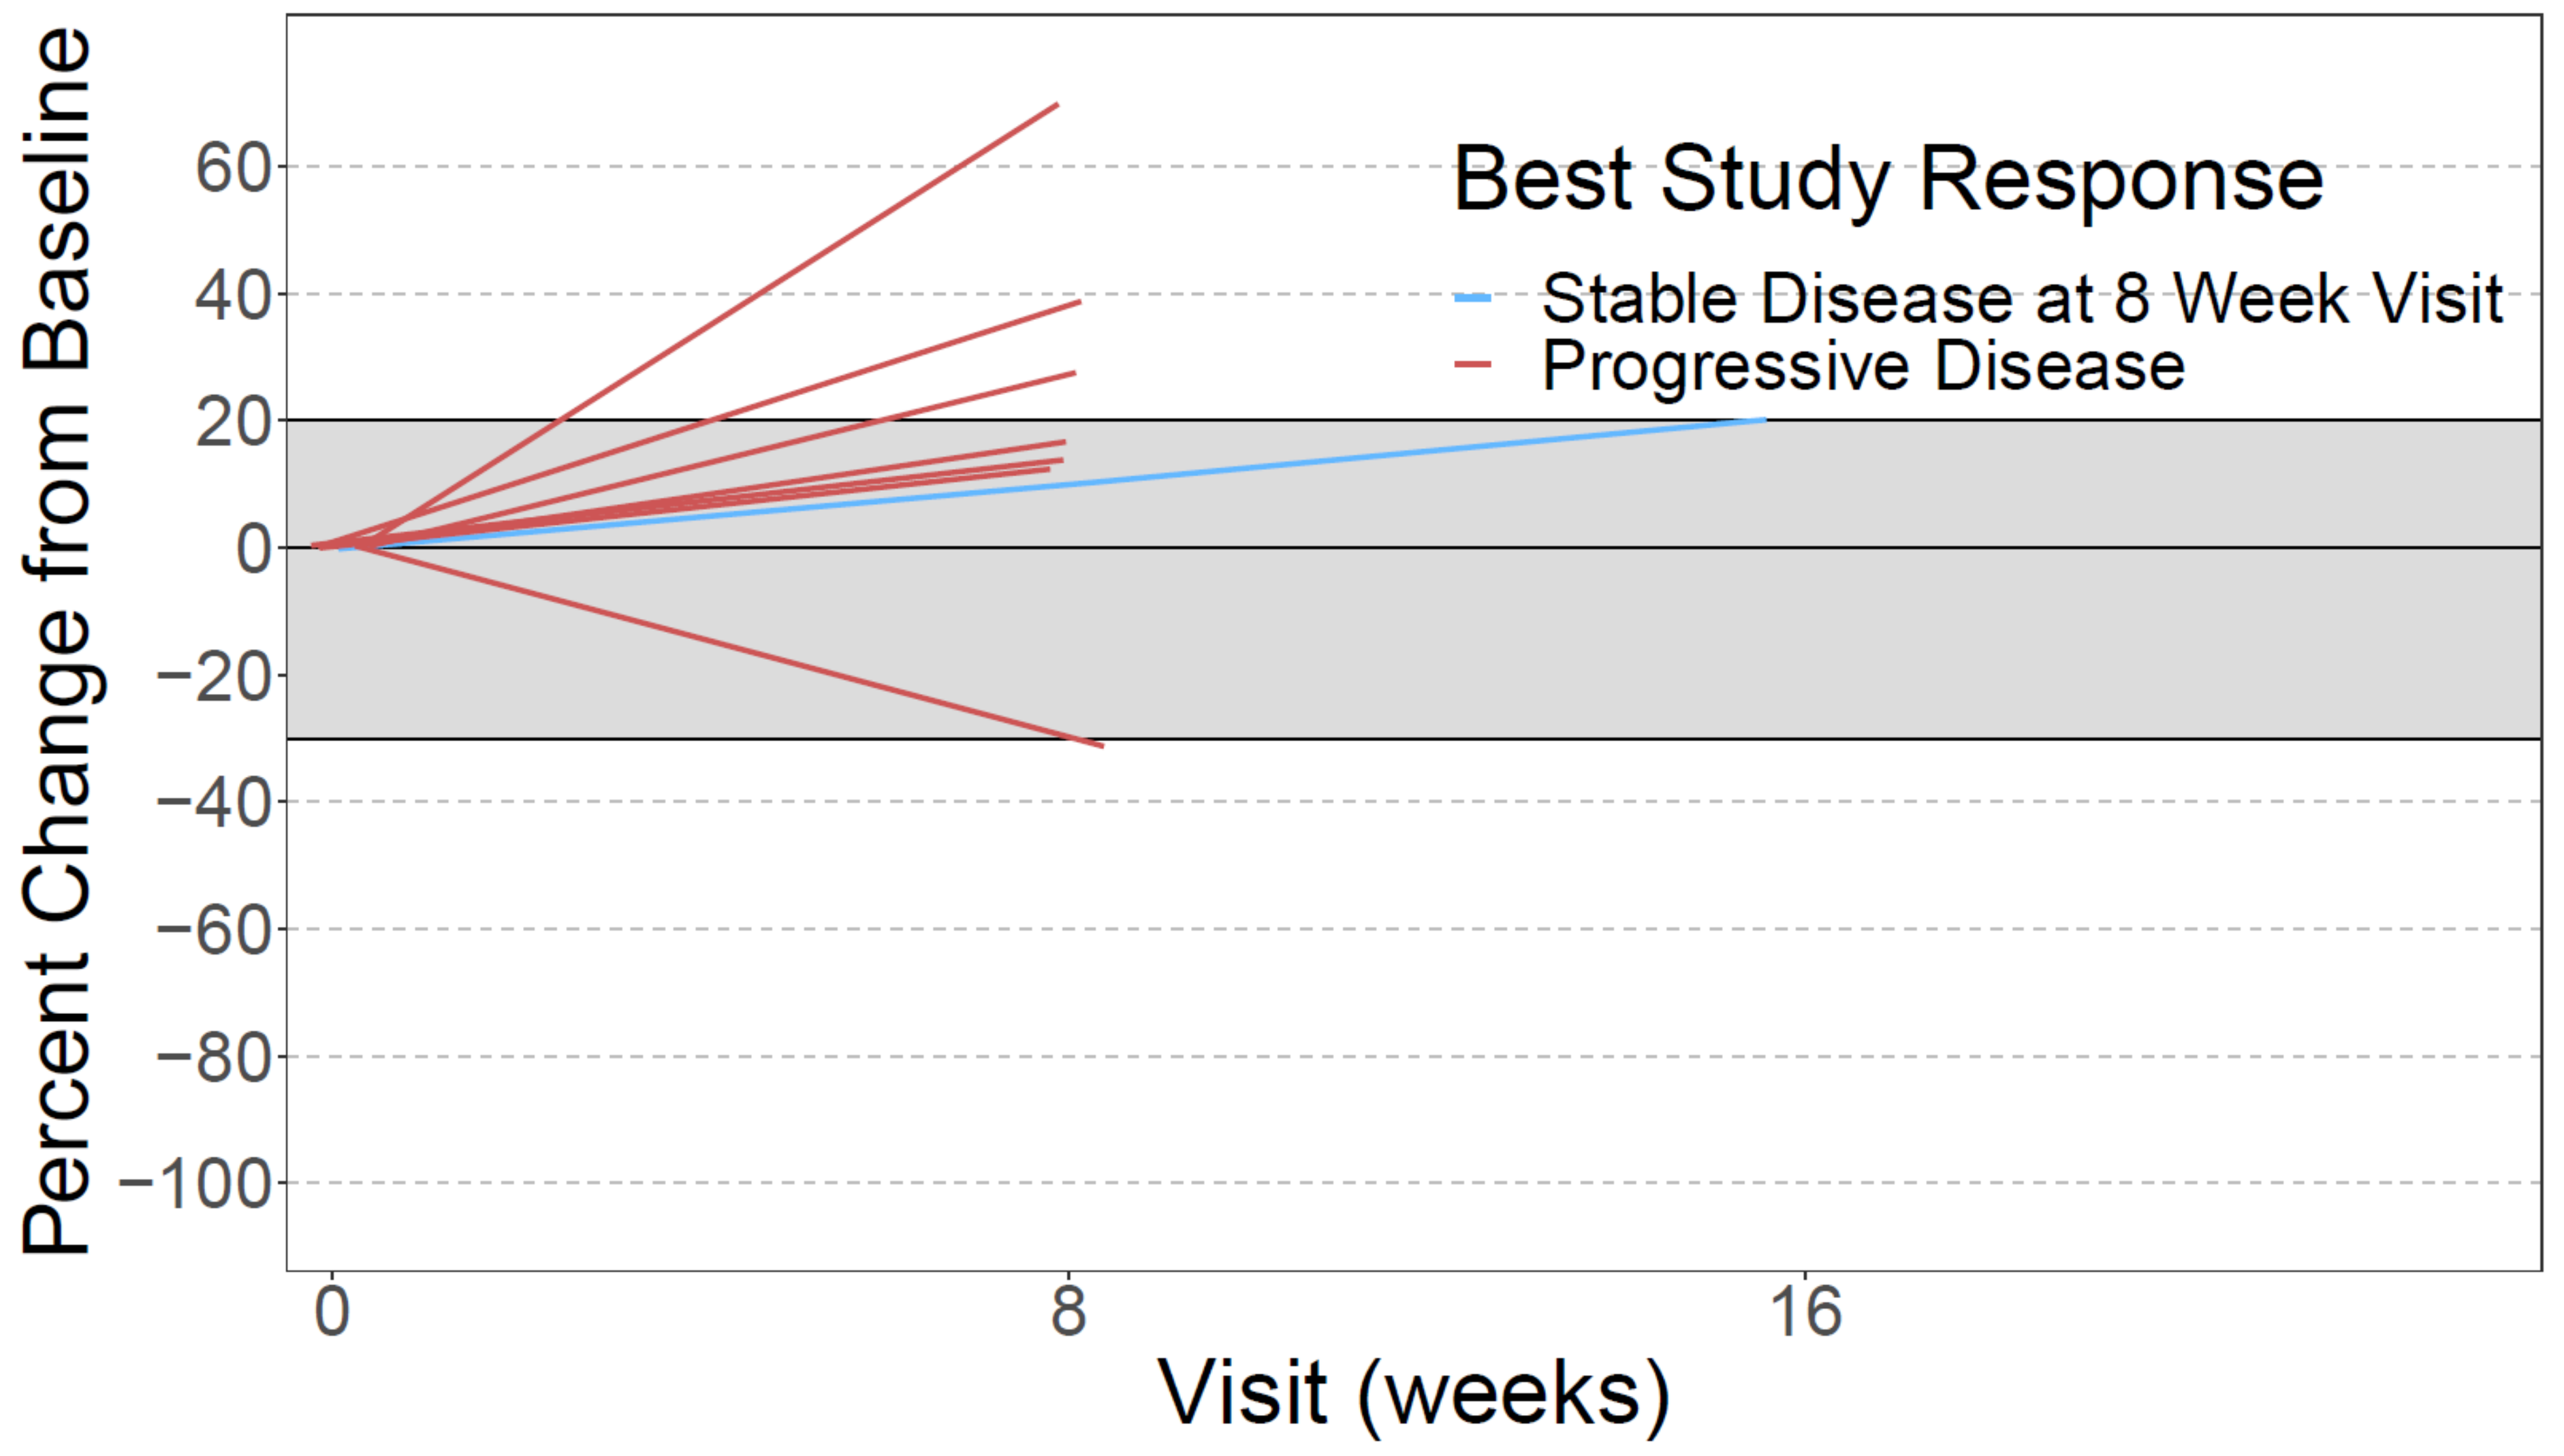
Percent Change from Baseline
60
40
20
0
−20
−40
−60
−80
−100
Best Study Response
Stable Disease at 8 Week Visit
Progressive Disease
0
8
16
Visit (weeks)